Targeted Delivery of siRNA to Renal Proximal Tubule Epithelial Cells Results in Approximately 70% Knockdown of Target Genes

Jeremy Cunniff, Shiying Ding, Johnny Lucas, Haojing Rong, Adam Belanger, Hongmei Zhang, Jonathan Lawrence, Andrew Fraley and Alfica Sehgal
Judo Bio, 300 Technology Square, Cambridge, MA 02139, USA

Background: Small interfering RNA (siRNA) is a clinically validated therapeutic modality, which silences gene expression via RNA interference (RNAi). Although siRNAs are secreted through the kidney, siRNA mediated knock-down in the kidney is still limited, largely due to challenges with optimal delivery. Proximal tubule epithelial cells (PTECs) within the nephron are attractive targets for utilizing RNAi, where the primary mode of entry is likely endocytic uptake and exposure is accessed through renal clearance of oligonucleotides. Here we describe a targeted approach to deliver conjugated siRNAs exploiting the PTECs' internalizing and recycling receptors.

We are developing our platform, STRIKE (Selectively Targeting RNA Into KidnEy) platform, that uses a proprietary approach to create ligand-RNA conjugate drugs designed for receptor-mediated update by specific kidney cell types, resulting in gene silencing of disease-modifying target genes.

In the present study, we investigated delivery of ligand conjugated siRNA, also called Megalin STRIKERs, and compared them to naked siRNA in mouse. These ligands were designed to specifically target proximal tubules, via the megalin receptor family. With different ligands, we achieved 5~30-fold increased exposure in kidney when compared to naked siRNA. These ligand conjugated siRNA led to enhanced gene knock-down specifically in the kidney.

In summary, conjugation with selected ligands of megalin receptor family, significantly enhanced siRNA uptake in mouse kidneys. In addition, these conjugations led to significant gene knock-down across multiple target genes.

Future work will focus on ligand conjugation optimization and chemical modification of oligonucleotides to improve kidney specific silencing activity.

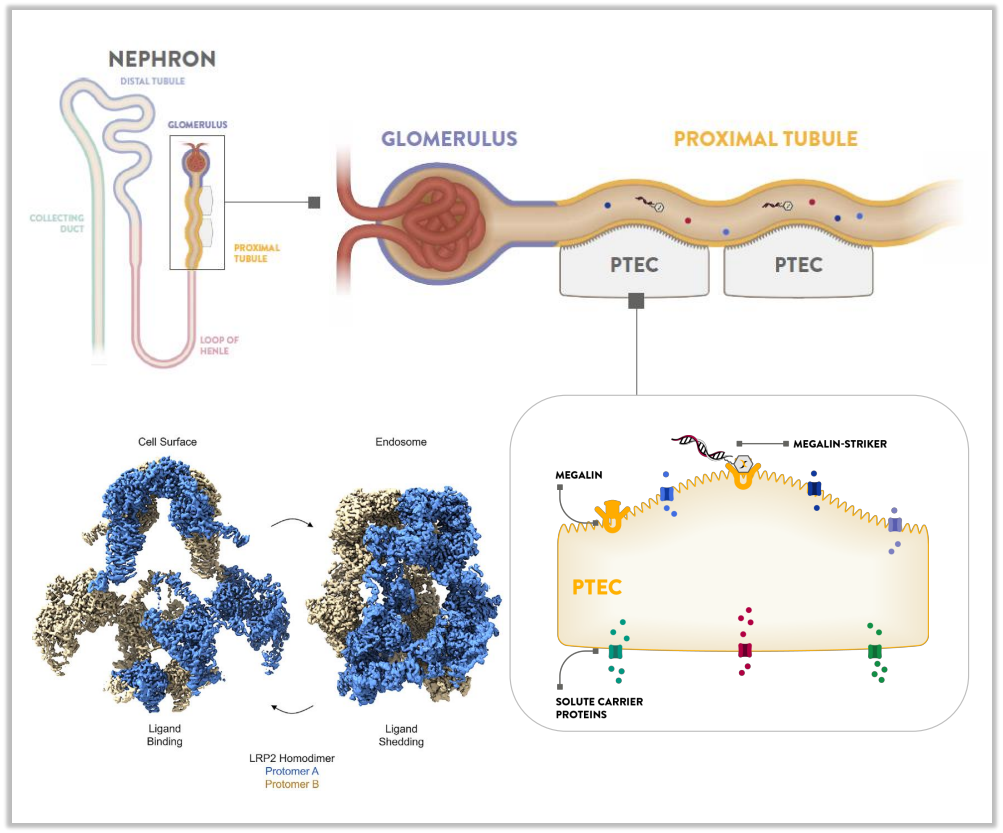
NEPHRON
DISTAL TUBULE
GLOMERULUS
COLLECTING DUCT
PROXIMAL TUBULE
LOOP OF HENLE
GLOMERULUS
PROXIMAL TUBULE
PTEC
PTEC
Cell Surface
Endosome
Ligand Binding
Ligand Shedding
LRP2 Homodimer
Protomer A
Protomer B
MEGALIN-STRIKER
MEGALIN
PTEC
SOLUTE CARRIER PROTEINS

Figure 1: Each kidney contains about one million nephrons, the functional unit of the kidney, that together filter about 50 gallons of blood each day. Blood flows through each nephron, beginning at the glomerulus then the proximal tubule on its way out via the collecting duct. Megalin is a cell-surface receptor that is highly expressed on proximal tubular epithelial cells (PTECs). It is rapidly internalized, slowly degraded, with high recycling capacity, making it an ideal entry point for intracellular delivery of a ligand-siRNA conjugate. Megalin structures refer to PDB code 8EM4 and 8EM7

Megalin ligands lead to increased knockdown of target mRNA compared to unconjugated siRNA

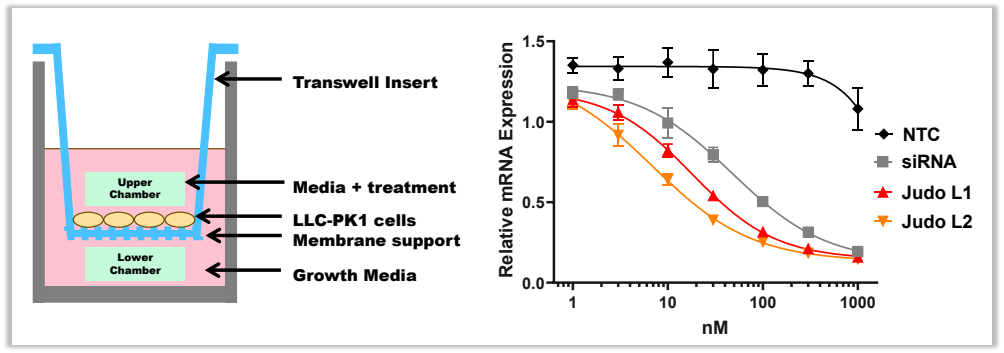
Transwell Insert
Upper Chamber
Media + treatment
LLC-PK1 cells
Membrane support
Lower Chamber
Growth Media
Relative mRNA Expression
nM
NTC
siRNA
Judo L1
Judo L2

Figure 2: LLC-PK1 cells were seeded on Transwell supports and treated apically for 3 hours with either unconjugated or Megalin ligand conjugated siRNA (Judo Ligand-1 & Judo Ligand-2, two classes of ligands). Cells were harvested 24 hours after treatment. The megalin-ligand conjugation enhanced the knockdown of the target gene compared to the unconjugated siRNA ($IC_{50:}$ unconjugated – 47.6 nM, Judo L1 – 18.2 nM, Judo L2 – 6.8 nM).

Ligand conjugated siRNA led to 5-30X increased uptake, specifically in kidneys

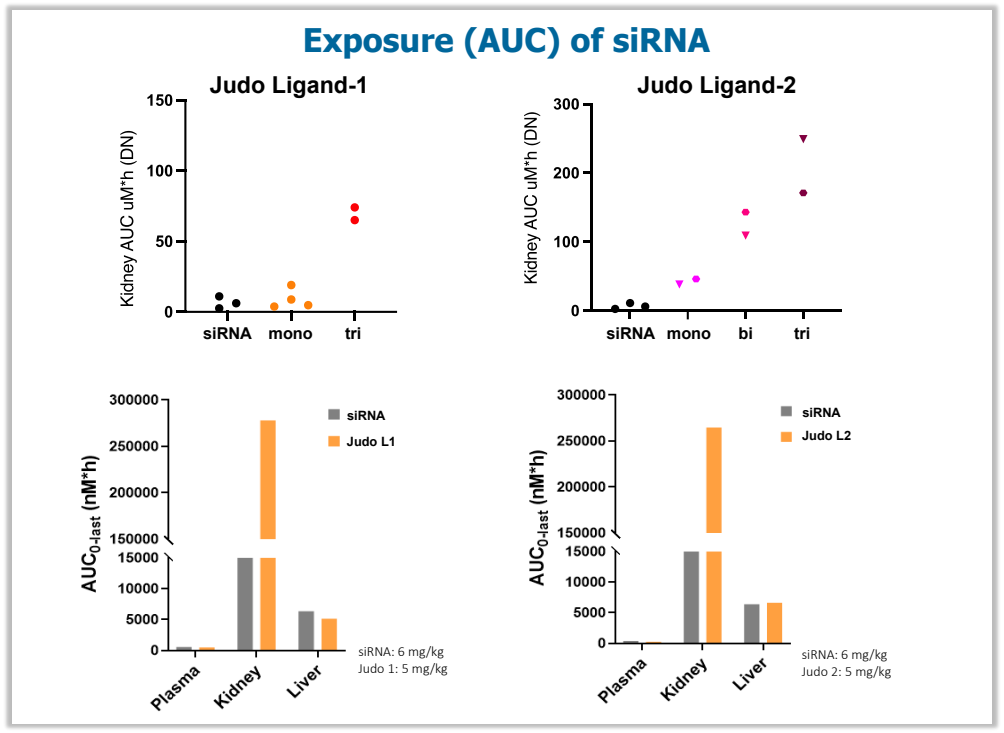
Exposure (AUC) of siRNA
Judo Ligand-1
Judo Ligand-2
Kidney AUC uM*h (DN)
siRNA
mono
bi
tri
AUC0-last (nM*h)
siRNA
Judo L1
Judo L2
Plasma
Kidney
Liver
siRNA: 6 mg/kg
Judo 1: 5 mg/kg
Judo 2: 5 mg/kg

Figure 3: Ligand conjugated siRNA families of Judo Ligand-1 and Judo Ligand-2 were tested in mice for tissue and plasma distribution. Mice were administered with a single dose of conjugated siRNA. Judo L1 and L2 were tested in multiple valencies attached to siRNA. Both Judo ligand conjugated to siRNA led to increase in kidney exposure over naked/unconjugated siRNA. This ligand mediated increase was specific to kidney and not seen in other tissues or plasma.

Megalin-STRIKERs have increased kidney uptake and targeted distribution to proximal tubules

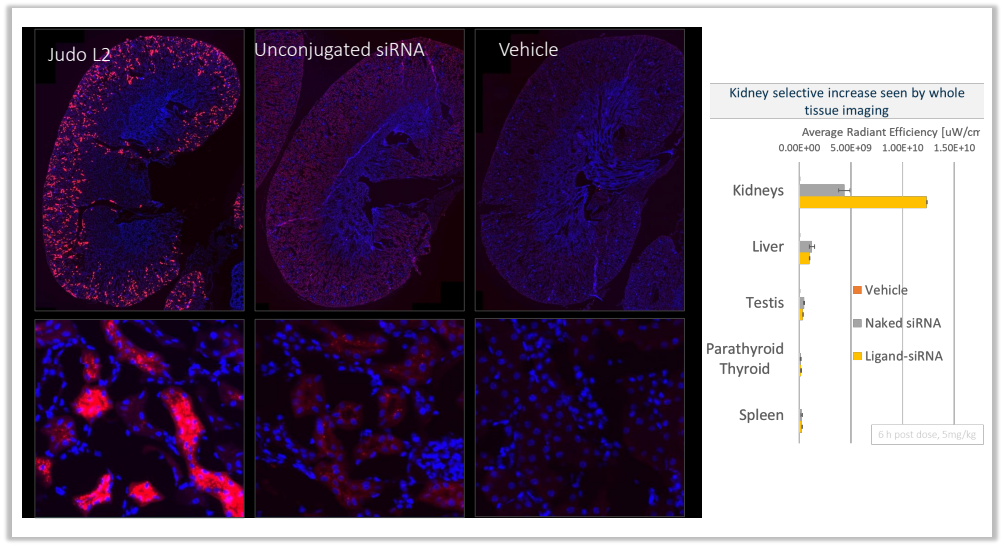
Judo L2
Unconjugated siRNA
Vehicle
Kidney selective increase seen by whole tissue imaging
Average Radiant Efficiency
Kidneys
Liver
Testis
Parathyroid Thyroid
Spleen
Vehicle
Naked siRNA
Ligand-siRNA

Figure 4: Mice were treated with Cy5.5-conjugated test articles at a dose of 5 mg/kg and sacrificed 6 hours after administration. Fluorescence associated with different organs was quantified and shown in the graph; Tissues were sectioned for fluorescent imaging. Judo-conjugated siRNA enhanced uptake in the kidney, specifically within PCTs. This enhanced uptake of conjugated siRNA was specific to kidneys, and not seen in other tissues.

Megalin STRIKERs mediated gene knock-down in mice

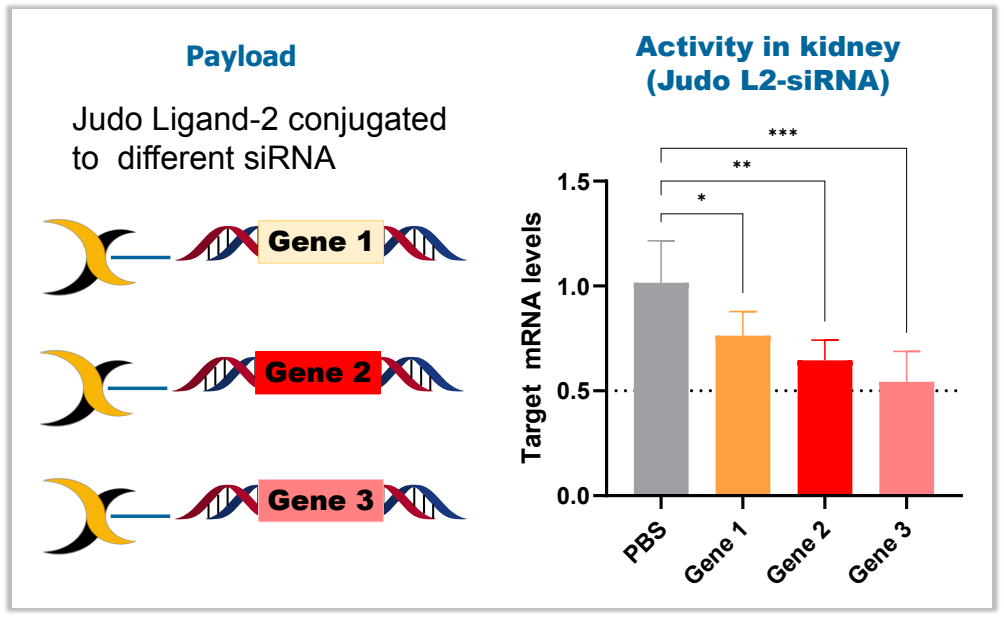
Payload
Judo Ligand-2 conjugated to different siRNA
Gene 1
Gene 2
Gene 3
Activity in kidney (Judo L2-siRNA)
Target mRNA levels
PBS
Gene 1
Gene 2
Gene 3

Figure 5: Judo Ligand-2 family ligands were conjugated to siRNA targeting three different gene targets. Mice (n=4 per group) were injected subcutaneously at 10ul/g and necropsied on Day 7. A knockdown of approximately 30~50% was achieved in the kidneys across different mRNA targets. The STRIKE platform is modular, allowing for the same ligand linker to be used across different siRNA.

~70% durable target gene knockdown attained in mouse kidneys with megalin-STRIKERs

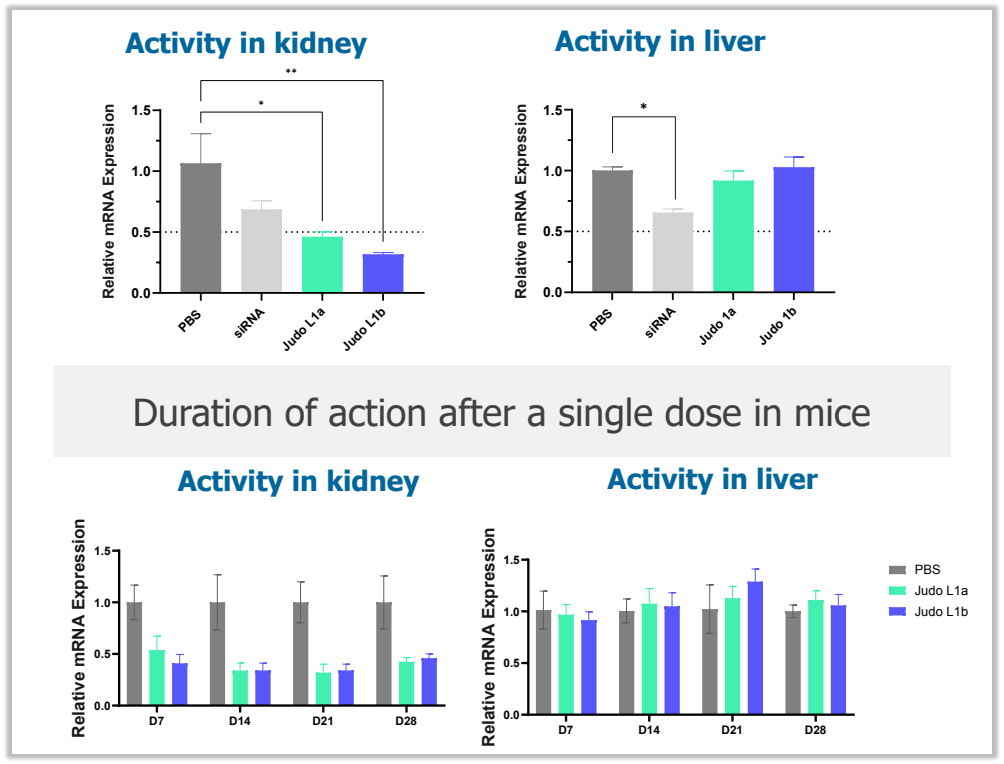
Activity in kidney
Activity in liver
Relative mRNA Expression
PBS
siRNA
Judo L1a
Judo L1b
Judo 1a
Judo 1b
Duration of action after a single dose in mice
Activity in kidney
Activity in liver
D7
D14
D21
D28
PBS
Judo L1a
Judo L1b

Figure 6: Mice (n=4 per group) were administered a subcutaneous dose of two variants of Judo Ligand-1 (Judo L1a and L1b). Significant knockdown of target gene was observed in the kidney at day 7, with greater knockdown of variant L1b. These ligand conjugated siRNA were ineffective in liver, consistent with lack of megalin expression in liver.

Sustained knockdown, upto 4 weeks was monitored in animals after a single subcutaneous dose administered on day 0. Both L1a and L1b conjugated siRNA show durable knockdown of target gene, lasting upto 4 weeks, the last time point tested.

Conclusion

The STRIKE (Selectively Targeting RNA Into KidnEy) platform represents a significant advancement in targeted siRNA delivery for renal applications. By leveraging ligand-siRNA conjugates mediated by megalin receptor, we achieved a remarkable 5~30-fold increase in siRNA exposure in mouse kidneys compared to naked siRNA. This enhanced delivery facilitated significant gene knock-down across multiple target genes specifically in proximal tubule epithelial cells (PTECs). These results highlight the utility of our approach in optimizing gene silencing for therapeutic interventions in systemic and renal diseases. Future research will focus on refining ligand conjugation and chemical modifications to further enhance nephron cell specific silencing efficacy.